IRM in a Multi-Resistant Malaria Vector Scenario Mexico Trial

Insecticide Resistance Action Committee

Objectives

To establish whether predicted methods of resistance management, based on mathematical models, would work under operational conditions, the IRAC Public Health Team sponsored an ambitious resistant management program against the multi-resistant New World malaria vector, *Anopheles albimanus*

In the coastal plain of Chiapas, Mexico, a large scale field trial was undertaken from 1996-2002 to evaluate rotations and mosaics of insecticides. The site was chosen because of the extensive history of insecticide use in Mexico. Extensive agricultural and public health insecticide used during the 1960's and 1970's selected multiple insecticide resistance mechanisms in *Anopheles albimanus*, the main coastal malaria vector. Subsequent changes in land use, the reduction in cotton farming and the success of malaria control activities consequently decreased Insecticide use.

This resulted in a well-documented regression towards insecticide susceptibility in *Anopheles albimanus* to all insecticides except DDT – as measured by diagnostic WHO mortality tests (*see table below*). Pressure for DDT resistance was maintained by continued use of this insecticide for malaria control in the regions surrounding the trial site.

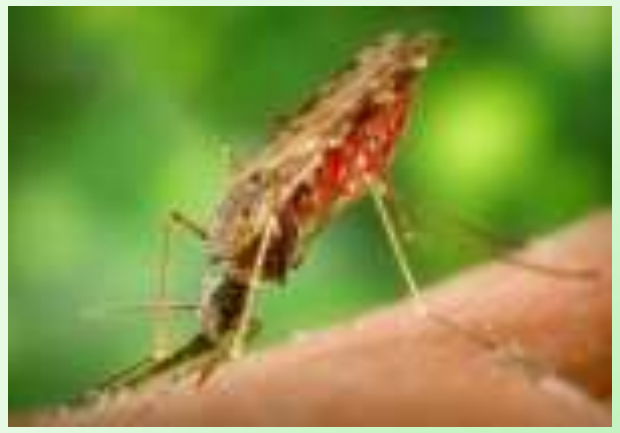

Adult Anopheles albimanus feeding

Mortality of *Anopheles albimanus*

Data from the Chiapas coastal plain to WHO diagnostic adult doses of different insecticides during the early 1980's and late 1990's.

Insecticide	Conc. (%)	1982	1983	1990	1997
DDT	4	38	39	47	40
Malathion	5	84	93	99	100
Fenitrothion	1	44	57	99	100
Fenthion	2.5	97	100		99
Chlorphoxim	4	98	99	100	10
Propoxur	.1	89	95		100
Deltamethrin	0.025	64	57	86	99
Cypermethrin	0.1		82		100
Bendiocarb	0.1		87		100
Pirimiphos methyl	4		99		100

The Study Design

Twenty four villages were selected and grouped into sets of three villages, which were randomly assigned to one of four treatment regimes

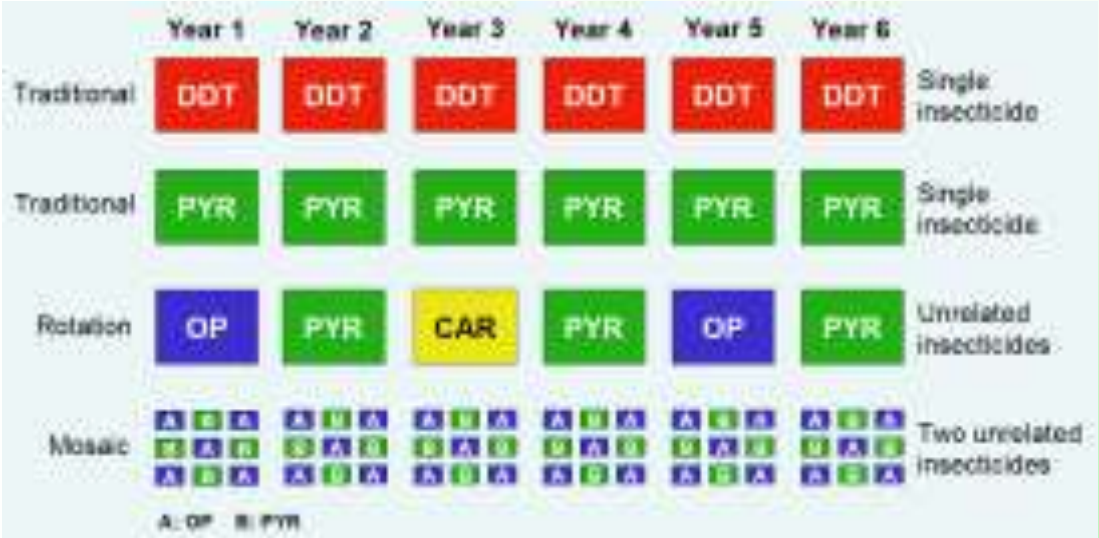

Year 1
Year 2
Year 3
Year 4
Year 5
Year 6
Traditional
DDT DDT DDT DDT DDT DDT
Single insecticide
Traditional
PYR PYR PYR PYR PYR PYR
Single insecticide
Rotation
OP PYR CAR PYR OP PYR
Unrelated insecticides
Mosaic
Two unrelated insecticides
A: OP B: PYR

Location – Chiapas, Mexico

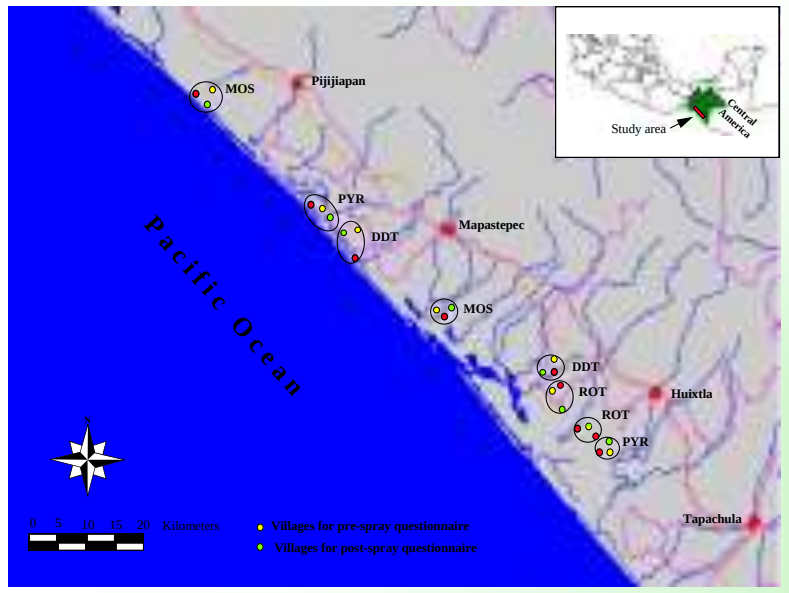

MOS
Pijijiapan
PYR
DDT
Mapastepec
MOS
Pacific Ocean
DDT
ROT
ROT
PYR
Huixtla
Tapachula
Study area
Central America
0 5 10 15 20 Kilometers
Villages for pre-spray questionnaire
Villages for post-spray questionnaire

STUDY AREA

Showing groups of villages & the treatments assigned.

MOS mosaic application,
PYR pyrethroid single use
DDT DDT single use
ROT annual rotation of insecticides

Treatments

All insecticides involved in the study were applied as part of normal anti-malarial activities three times per year, with the exception of DDT, which was sprayed twice per year. Insecticides were sprayed with a Hudson X-Pert® sprayer with nozzle No. 8002.

Wall bioassays to monitor residual efficacy of insecticides were conducted one day and then every month, after spraying. Good killing effect of mosquitoes was achieved with all products at the applied dosages (OP at 2 g a. i./m2, Pyrethroid at 0.025 g a. i./m2, Carbamate at 0.4 g a. i./m2 and DDT at 2 g a.i./m2), with mosquito mortalities averaging around 75% four months after insecticide application.

Field-Caught Mosquitoes in the Lab

The frequency of all resistance mechanisms was monitored before and during the intervention period by biochemical assays, along with WHO diagnostic bioassays using insecticide impregnated papers. Field samples of mosquitoes were collected on a regular basis approximately three months after each spray round and the F1 generation reared from the field-caught mosquitoes were used for all assays. When few mosquitoes were available, priority was given to biochemical assays since this method was the most sensitive for detection of small changes in resistance. Biochemical assay results were compared with the *Anopheles albimanus* susceptible Panama strain. Logistic regression analyses were used to determine the effect of the different treatment regimes on the frequency of different resistance mechanisms.

Pyrethroid treatment and pre-spray were set as reference variables in the analysis. Since no changes were observed in DDT resistance levels under any treatment regime during the whole study period, data from DDT treated villages were excluded from the analyses.

Rotation or Mosaic Schemes More Effective

Bioassays showed that continuous use of a pyrethroid gradually increased pyrethroid resistance in the mosquito field population over the first four years: resistance then remained stable for the next two years. In the rotation and mosaic schemes, pyrethroid and organophosphate resistance were selected at low levels and remained stable. No carbamate resistance was observed in the rotation scheme.

The biochemical assays showed that although enzymes activity patterns varied, the chances of high level resistance development using a rotation or a mosaic regime were significantly lower than the rate at which resistance was selected using a pyrethroid alone.

Managing Insecticide Resistance or Avoiding its Evolution

Both the rotation and mosaic strategies performed well in the trials, however the practicality of operating a mosaic scheme may pose too many logistical difficulties in a real control programme. It would mean two different insecticides, two handling and dilution requirements, two different application rates!. It would also require records kept of which houses were treated with which insecticide. In terms of practicality it may slow applications too much to be realistic. hence the best and most practical option is the use of rotation schemes.

To operate rotation schemes the susceptibility status of the local mosquito population should be known before commencing and then monitored annually to pick up any changes or trends. The insecticides used should not be ones to which resistance or cross resistance already exists, although they may be re-introduced after several years when levels of susceptibility to them increase.

Most insecticides should be used on a minimum of a three year rotation. Consideration should also be given to integrating larviciding with different actives such as IGR`s or biologicals which have totally different modes of actions to the chemical adulticides.

This poster is for educational purposes only. Details are accurate to the best of our knowledge but IRAC and its member companies cannot accept responsibility for how this information is used or interpreted. Advice should always be sought from local experts or advisors and health and safety recommendations followed.

Information taken from an article in Bayer Public Health Journal 18/2006. Authors:A. D. RODRIGUEZ R. P. PENILLA, M. H. RODRIGUEZ J. HEMINGWAY

www.irac-online.org

Designed and produced by the IRAC Public Health Team, September 2007. Photograph courtesy of J. Gathany, CDC